Altered iNKT Cell Populations in the Airways of Patients With IPF

A. Agro[1], V. Kumar[1], A. Byrne[2], T. Maher[3], P. L. Molyneaux[2], E. Calamita[2], M. Hertz[1]

[1]GRI Bio Inc, San Diego, CA, United States, [2]Imperial College London, London, United Kingdom, [3]University of Southern California, Los Angeles, CA, United States

Introduction

Emerging evidence suggests that type 1 invariant NKT (iNKT) cell-associated mechanisms contribute to type 1, type 2, and type 3 immune pathways mediating tissue fibrosis, including IPF[1]. Supporting data contend that iNKT cells are at the proximal end of this pathway. As such, targeting a cell population upstream of these various immune mechanisms may be important in modulating various fibrotic conditions in a multifaceted manner.

Here we show that iNKT cells are present in increased number and activity in BAL from patients with IPF; that in the bleomycin-induced fibrosis model in mice, a selective inhibitor of iNKT cells, GRI-0621, can modulate the fibrotic condition and can reduce the important inflammatory and fibrotic drivers of the disease. Finally, the design of the Phase 2a study examining the safety, tolerability, and effect on various biomarkers of GRI-0621 is presented.

Background

NKT cells are a family of unconventional T cells with diverse functions. They bridge adaptive and innate immune responses and express both NK cell receptors and antigen T cell receptors (TCR)[1]. Human iNKT cells are effector T cells that mostly express the semi-invariant germ line encoded Vα24Jα18 paired with a more diverse non-germ line Vβ11 and can be identified with a clonotypic TCR-specific mAb. iNKT cells become rapidly activated following either TCR recognition of a CD1d-bound lipid antigen or inflammatory cytokine-associated signaling and produce large amounts of type 1, type 2, or type 3 cytokines. Accordingly, NKT1 predominantly secretes GM-CSF and IFN-g, NKT2 secretes IL-4, IL-5, and IL-13, and NKT17 secretes IL-17A and IL-22[1].

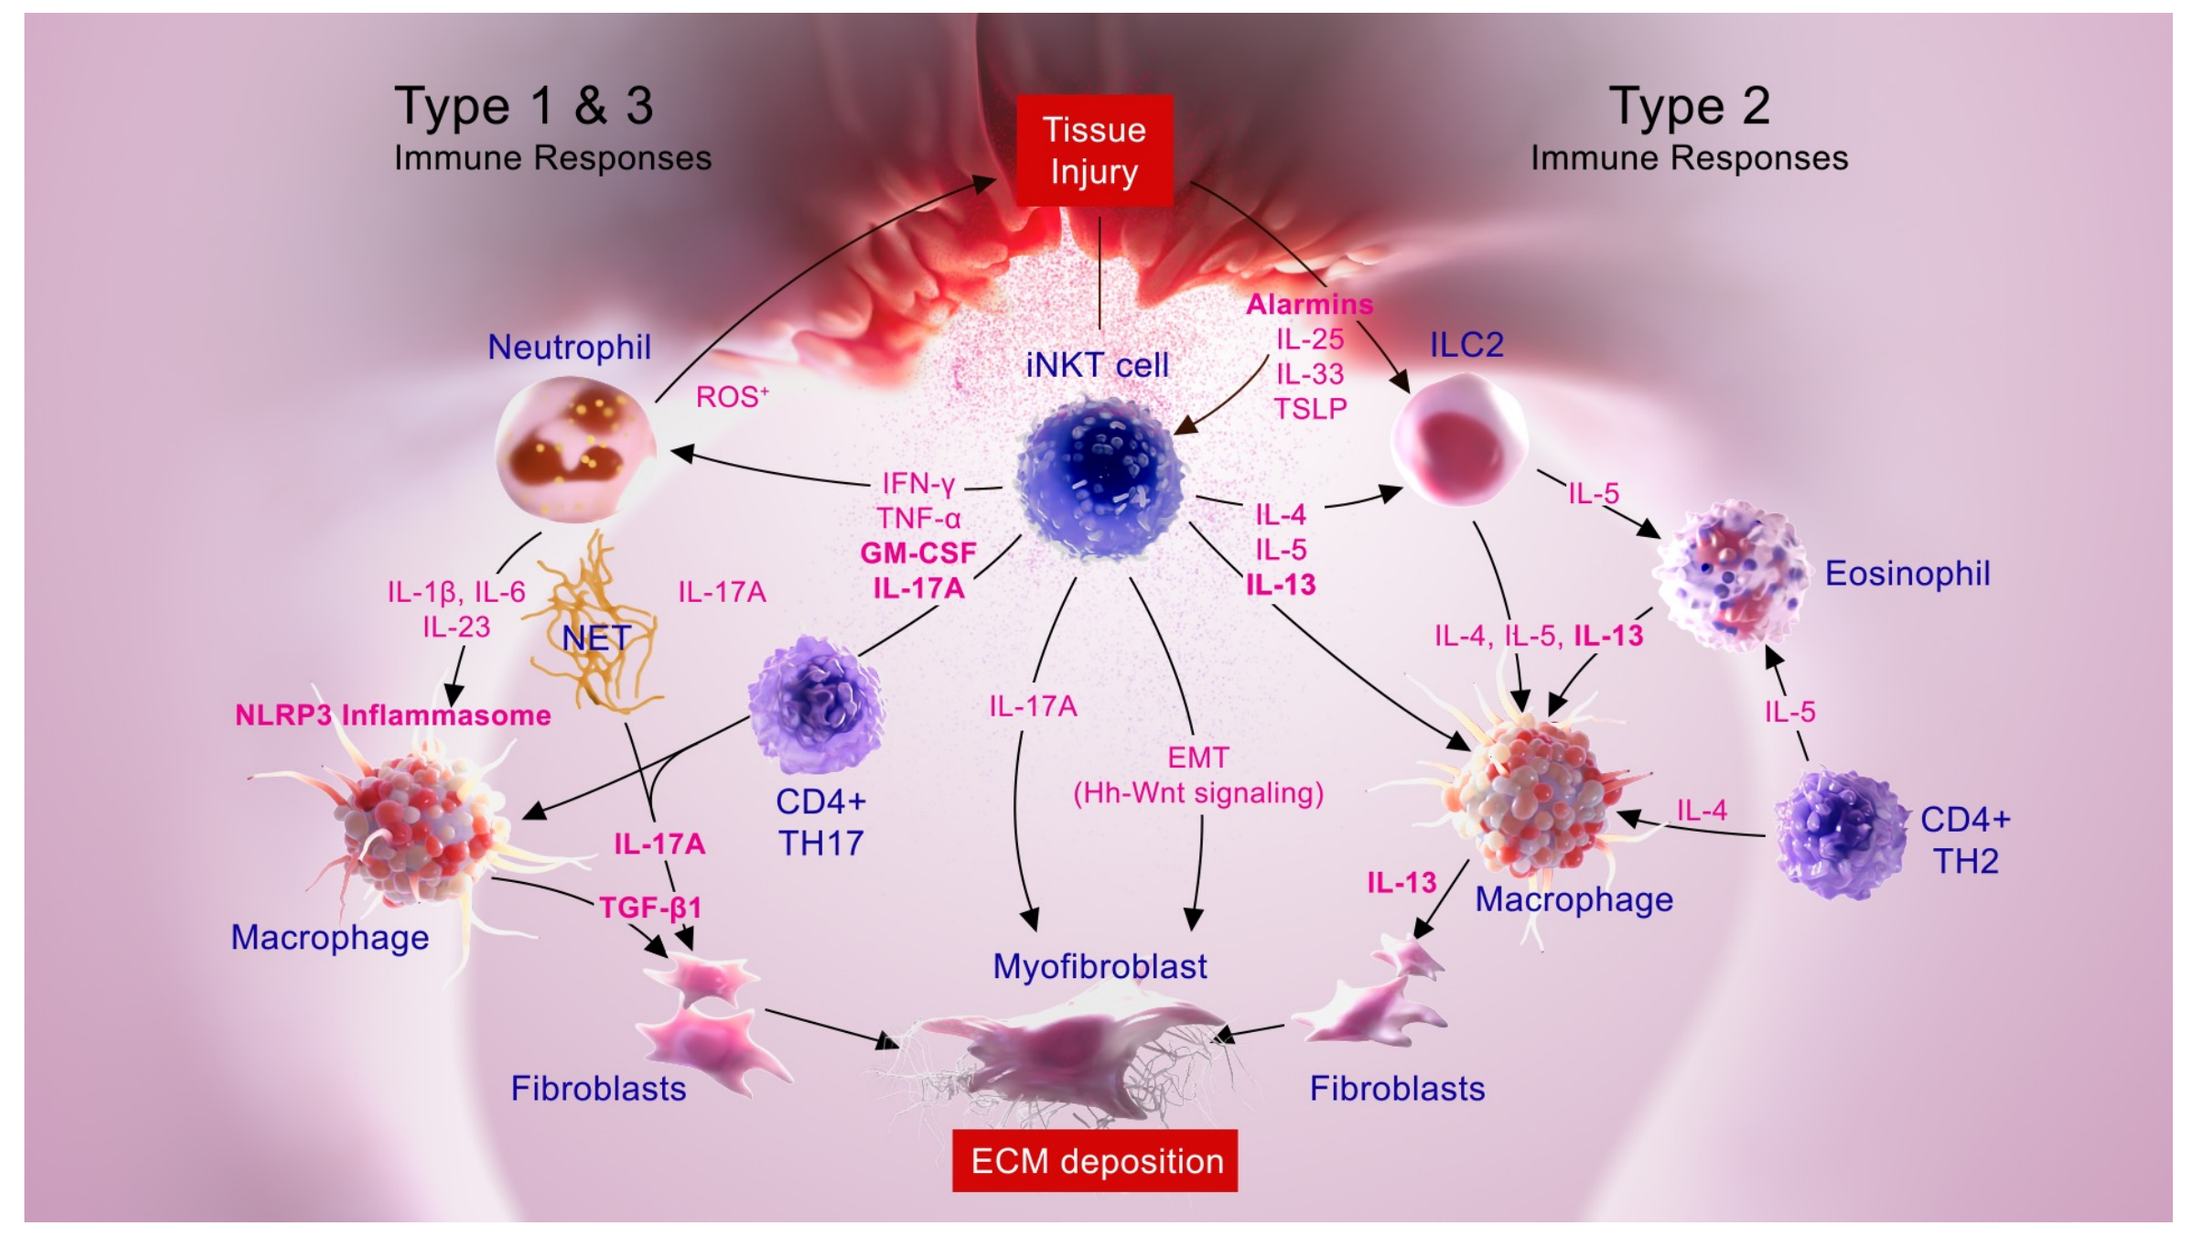
Type 1 & 3
Immune Responses
Type 2
Immune Responses
Tissue Injury
Neutrophil
ROS
iNKT cell
Alarmins
IL-25
IL-33
TSLP
ILC2
IFN-γ
TNF-α
GM-CSF
IL-17A
IL-4
IL-5
IL-13
IL-5
Eosinophil
IL-1β, IL-6
IL-23
NET
IL-17A
NLRP3 Inflammasome
CD4+
TH17
IL-17A
EMT
(Hh-Wnt signaling)
IL-4, IL-5, IL-13
IL-5
IL-4
CD4+
TH2
IL-17A
TGF-β1
IL-13
Macrophage
Macrophage
Myofibroblast
Fibroblasts
Fibroblasts
ECM deposition

iNKT cells in IPF patients

To investigate the role of pulmonary iNKT populations in IPF, we first analyzed bronchoalveolar lavage (BAL) fluid from IPF patients (n=129) and healthy controls (n=23), by quantitative PCR and by flow cytometry. IPF patients had increased expression in whole BAL pellets of Vα24-Jα18+ iNKT cells and factors known to be involved in lung fibrosis such as Collagen 1-α1, osteopontin, and TGF-β, as assessed by qPCR (see Fig. 1). There were no changes in the expression of TNF, CXCR3, IL-4, IL-5, or IL-13 in BAL pellets from IPF patients compared to healthy controls (data not shown).

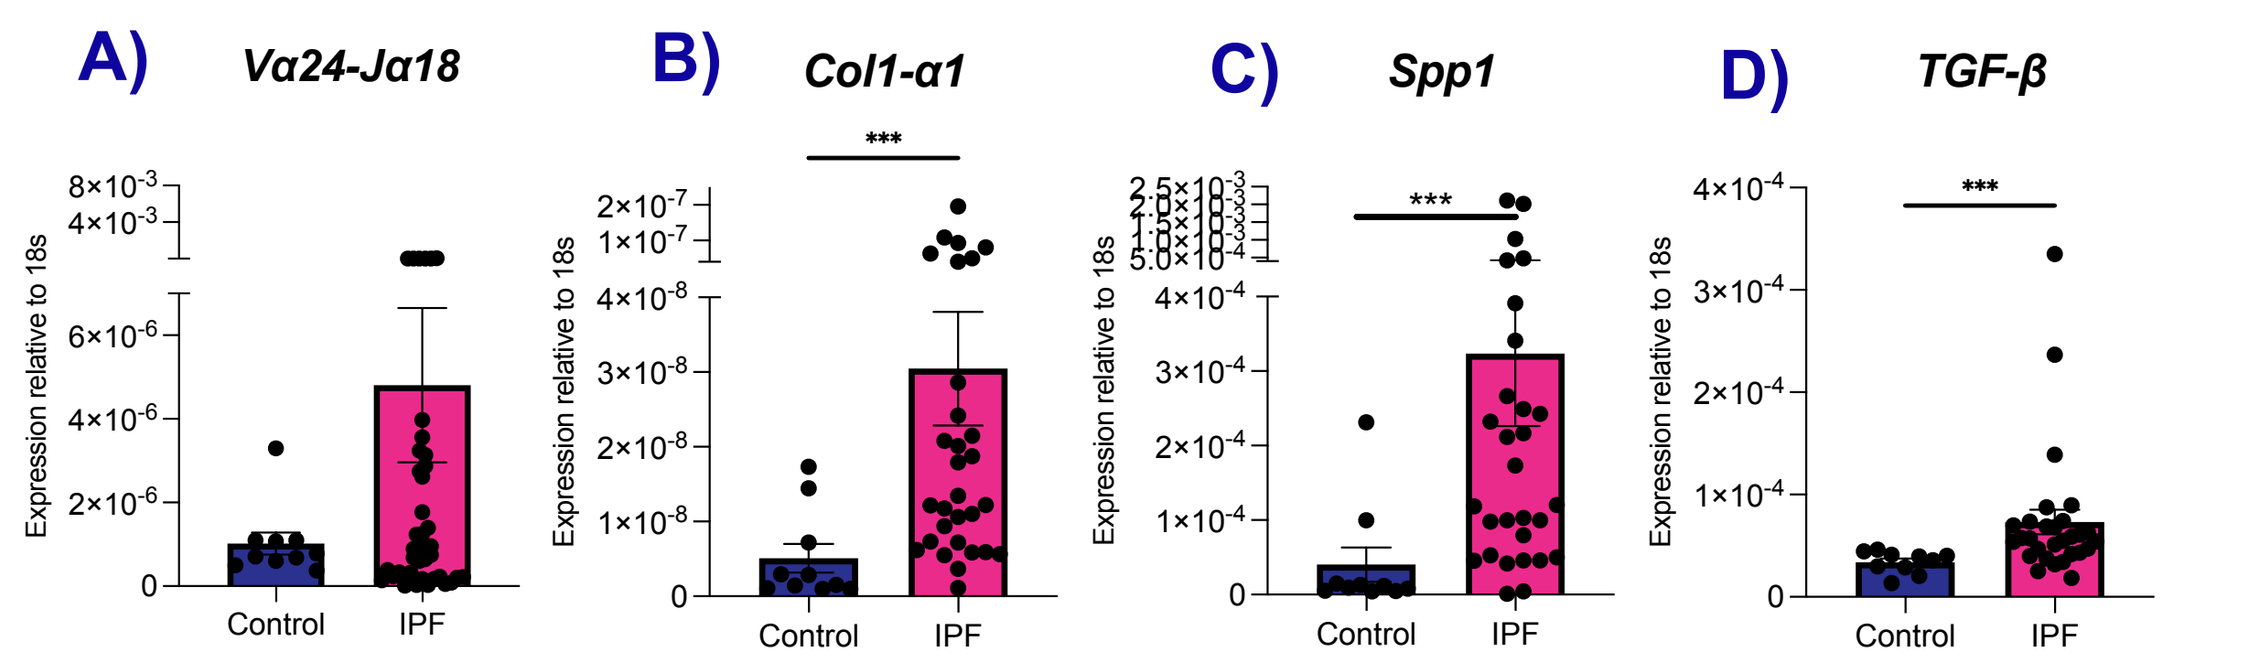
A)
Vα24-Jα18
B)
Col1-α1
C)
Spp1
D)
TGF-β
Expression relative to 18s
Control
IPF

Figure 1. Expression of A) Vα24-Jα18 B) Col1-α1, C) Spp1/osteopontin, or D) TGF-β in whole BAL pellets in healthy controls (n=10) or IPF patients (n=29). Data presented as mean ± SEM. * p<0.05, ** p<0.01, *** p<0.005

A trend towards increased expression of the iNKT TCR Vα24-Jα18 CDR3 region (Fig. 1A), suggested increased numbers of iNKT cells in the IPF BAL, compared to healthy controls. To further examine the iNKT cell signature in IPF, labeled cells were acquired on an LSR Fortessa (BD Biosciences).

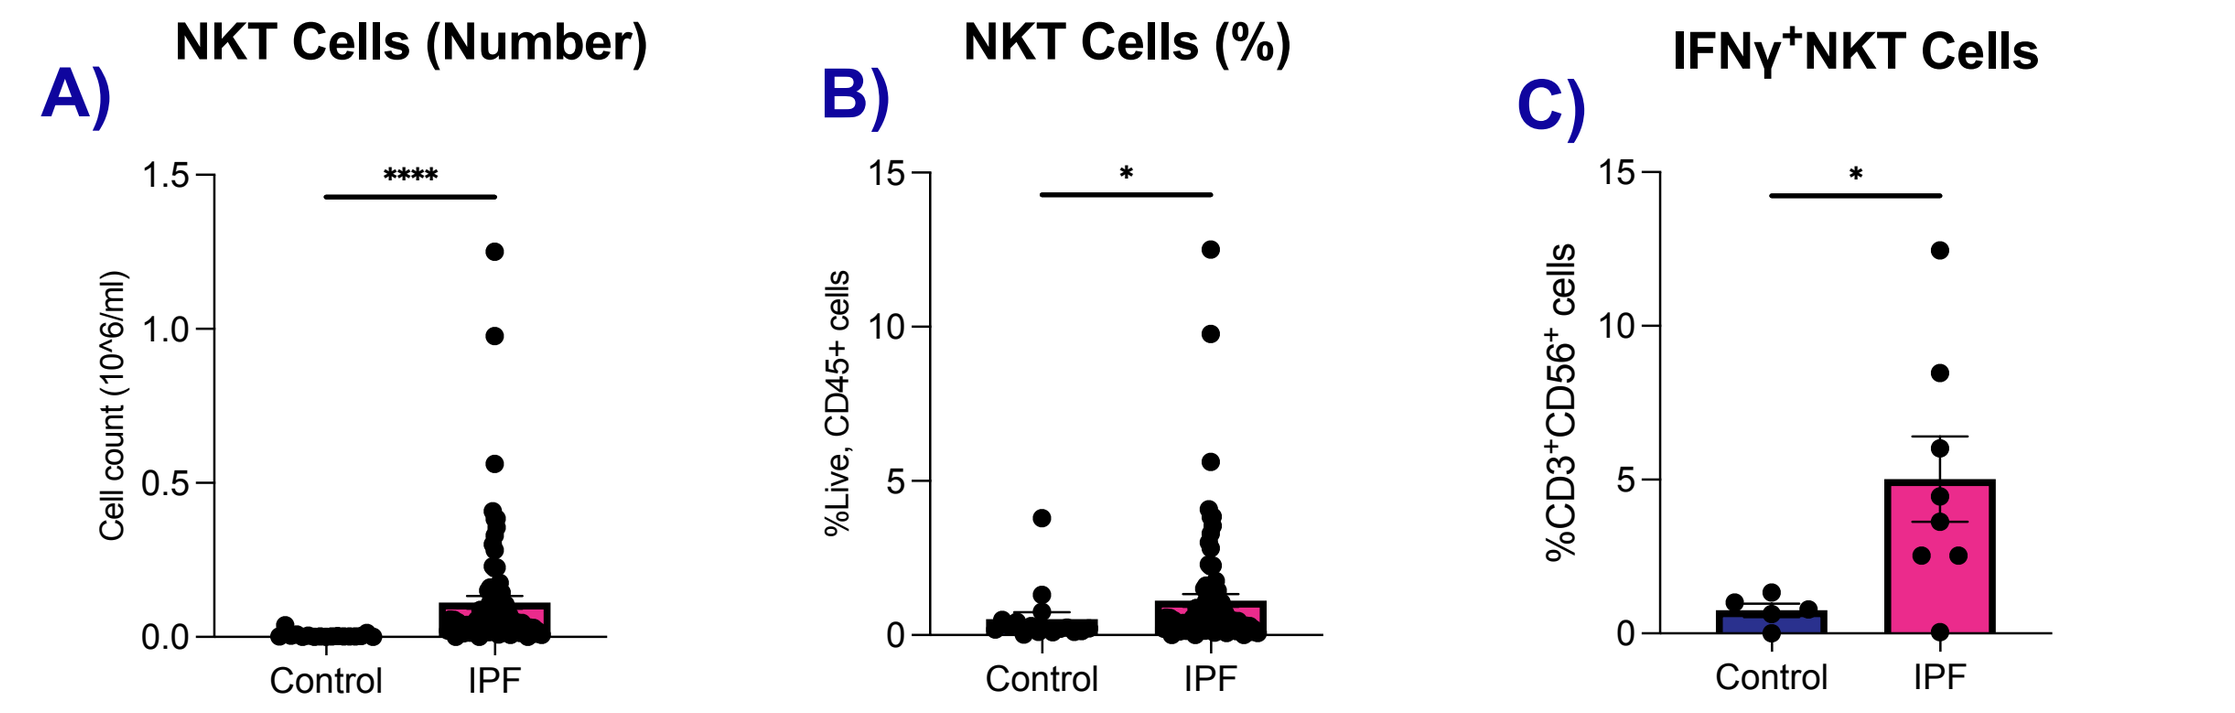
A)
NKT Cells (Number)
Cell count (10^6/ml)
B)
NKT Cells (%)
%Live, CD45+ cells
C)
IFNγ+NKT Cells
%CD3+CD56+ cells
Control
IPF

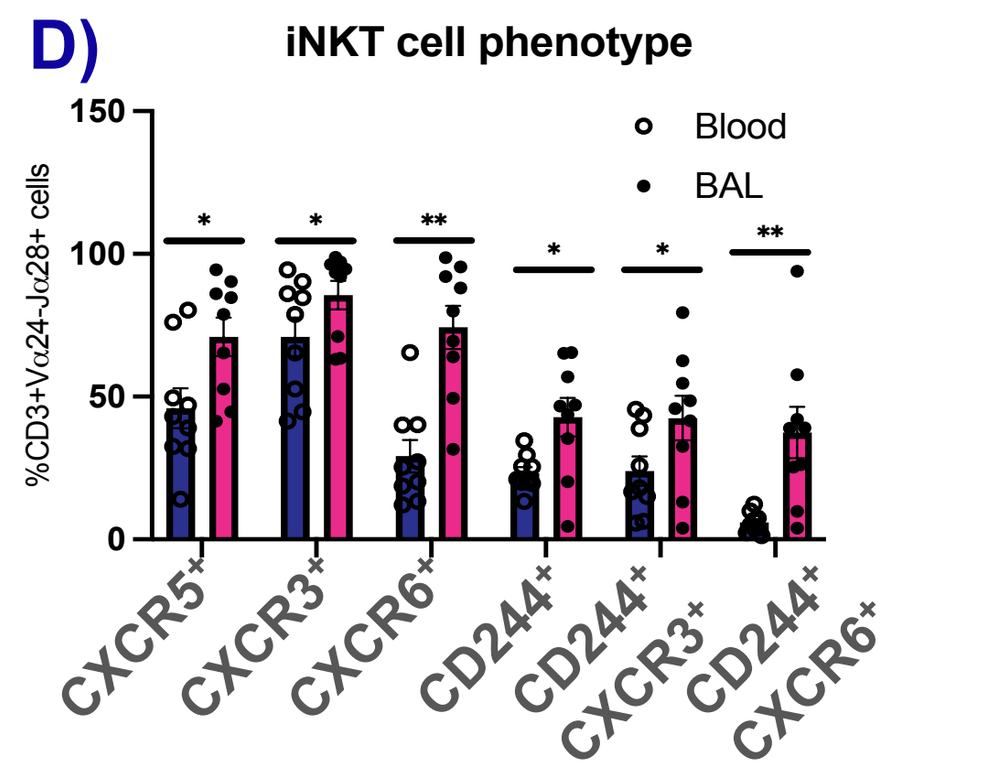
D)
iNKT cell phenotype
Blood
BAL
%CD3+Vα24-Jα28+ cells
CXCR5+
CXCR3+
CXCR6+
CD244+
CD244+ CXCR3+
CD244+ CXCR6+

Figure 2. Total numbers (A) and percent (B) iNKT cells in BAL of healthy (n=17) or IPF patients (n=86). IFN-γ+ iNKT cells (C) in BAL of healthy (n=5) or IPF patients (n=8). iNKT cells were identified as CD45+ CD3+ CD56+ cells. Data presented as mean ± SEM. * p<0.05, ** p<0.01, *** p<0.005, **** p<0.001.

Our data indicate that there are distinct populations of CD45+CD3+CD56+ (NKT cells) in IPF-BAL. Interestingly, numbers (Figure 2A) and proportion (Fig. 2B) of NKT cells were increased in IPF airways compared to healthy controls. To assess the NKT cell activation, we performed intracellular staining for IFN–γ in unstimulated BAL cells. We detected an increase in IFN–γ producing NKT cells in IPF compared to controls (Fig. 2C). We next confirmed the NKT cell phenotype in a second cohort, using an antibody against Vα24-Jα18 of the iNKT TCR. iNKT cells were differentially activated in the airway compared to the blood. BAL iNKT cells had increased expression of CXCR5, CXCR6, CXCR3, and CD244. Additionally, the proportions of CD244+CXCR6+ iNKT cells were significantly increased in BAL compared to blood (Fig. 2D). Recently, a circulating CD244+ CXCR6+ IFN-γ producing iNKT cell subset has been described in mice and humans, accumulate in liver and drive fibrosis[2-5].

GRI-0621

GRI-0621 is a small molecule, orally-delivered, RARβγ selective agonist in development for the treatment of IPF. GRI-0621 binds to iNKT cells that have been shown to express RARβγ and can inhibit the number and activity of iNKT cells in vitro, in models of fibrotic disease[4], and in human iNKT cells in vivo (NCT02949375). GRI-0621 is presently being evaluated in a Phase 2a proof-of-concept biomarker study in patients with IPF (NCT06331624).

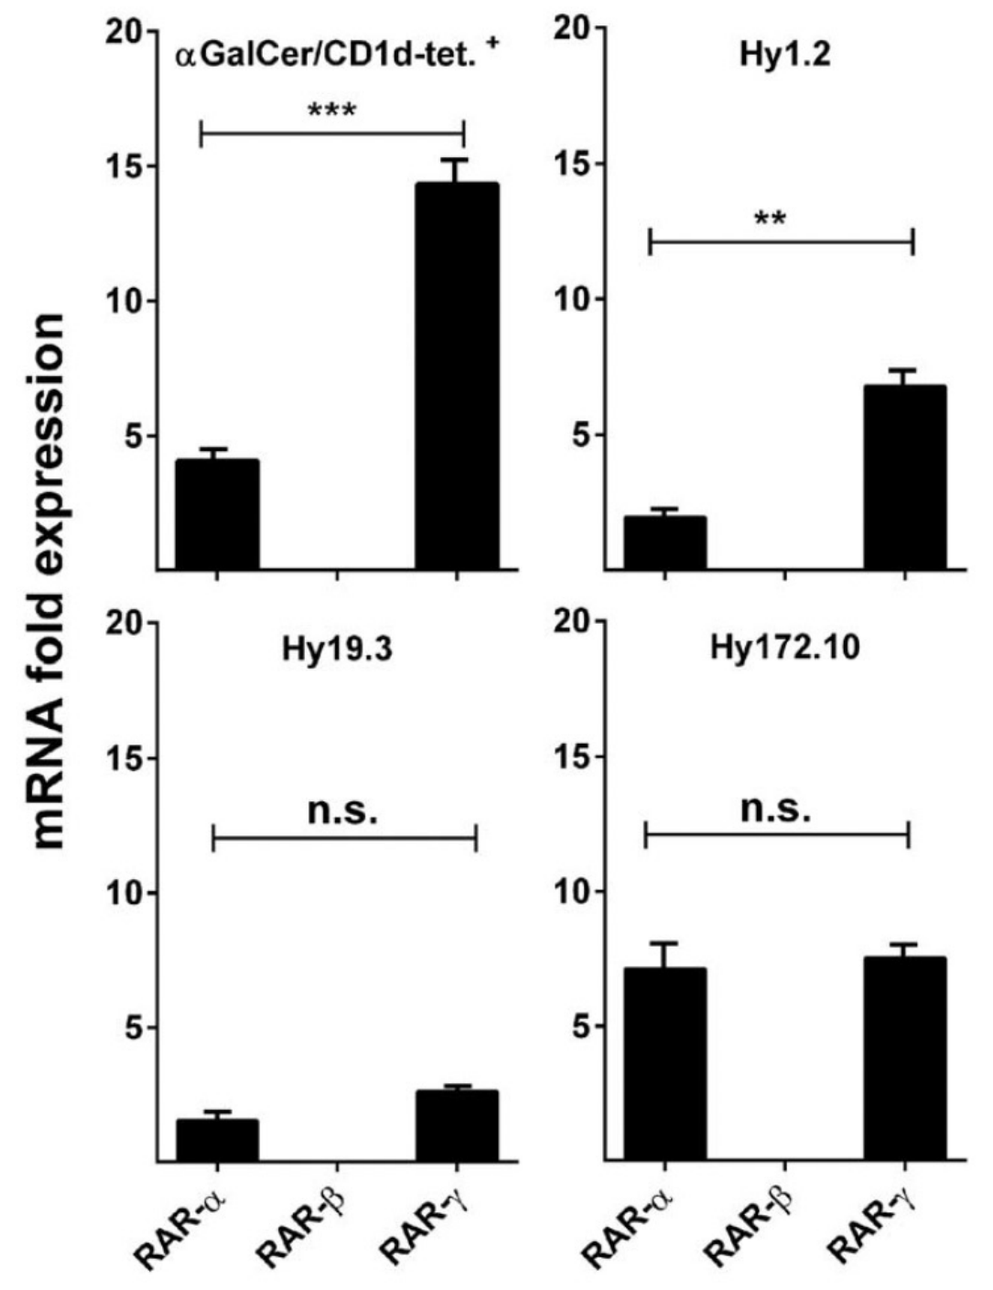
αGalCer/CD1d-tet.+
Hy1.2
Hy19.3
Hy172.10
mRNA fold expression
n.s.
RAR-α
RAR-β
RAR-γ

Figure 3. Constitutively high expression of RARγ in iNKT cells. Relative expression of RARα, RARβ, and RARγ genes using qPCR assay in a polyclonal population of iNKT cells or monoclonal populations of Hy1.2 (iNKT), Hy19.3 (type 2 NKT), and Hy172.10 (MBP-reactive) conventional CD4+ T cells is shown. Data representative of three independent experiments. ***P<0.001; **P<0.01.

iNKT activity in IPF model

To ascertain the functional role of NKT cells, we utilized the murine bleomycin model of pulmonary fibrosis. Wild-type (WT) C57BL/6 mice were instilled with a single dose of bleomycin 1.5 IU/Kg via the intratracheal route, with or without daily administration of GRI-0621 (1mM in PBS, a human equivalent dose), nintedanib (60 mg/kg), or vehicle control, via oral gavage (schematic below).

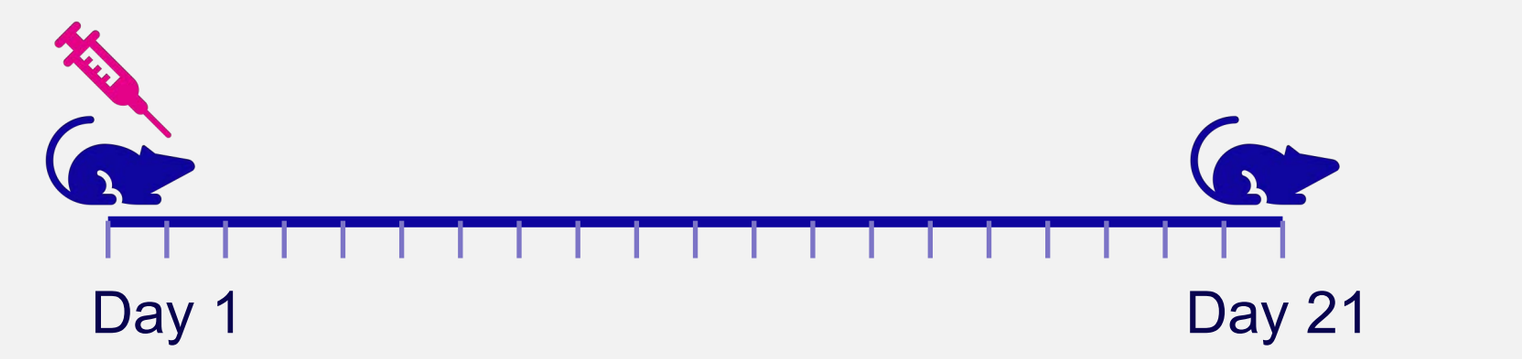
Day 1
Day 21

All animals (8-wk old C57BL/6 male mice), except those in the Sham group, were administered intratracheally bleomycin [1.5 IU/kg] on Day 1. Sham control mice received the same volume of saline IT with no bleomycin or treatment. Test groups received GRI-0621 (1 mg/kg) or Nintedanib (60 mg/kg) by oral gavage once daily for 21 days starting after bleomycin administration.

Compared to controls, mice treated with GRI-0621 showed improved fibrosis and immunopathology. Inhibition of iNKT activity led to a decrease in fibrosis score (Fig. 4A) and total lung inflammation. To determine the impact of iNKT cell inhibition on immune responses in the lung, we performed differential cell counts (automatic hematology analyzer, XN-1000 Sysmex, Japan). GRI-061 treated mice had reduced numbers of total WBC, neutrophils, monocytes, and lymphocytes (Fig. 4B and data not shown)

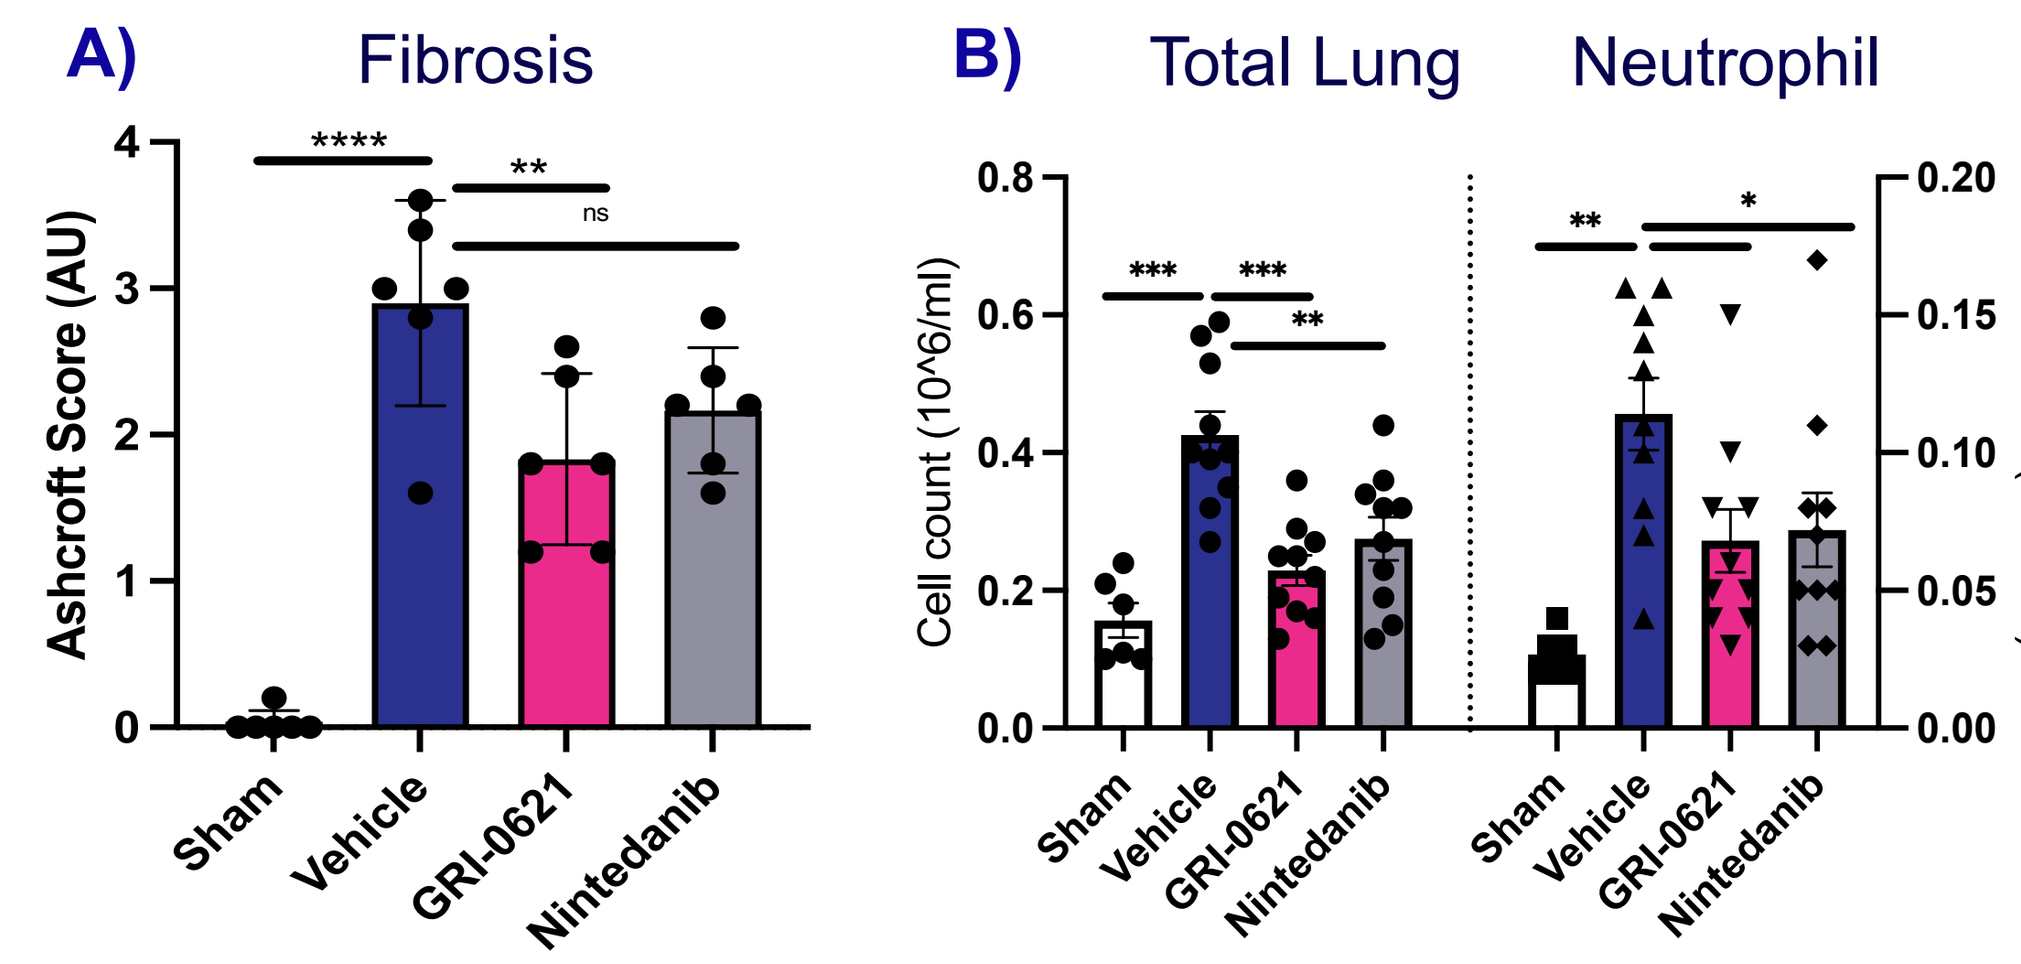
A)
Fibrosis
Ashcroft Score (AU)
ns
B)
Total Lung
Neutrophil
Cell count (10^6/ml)
Sham
Vehicle
GRI-0621
Nintedanib

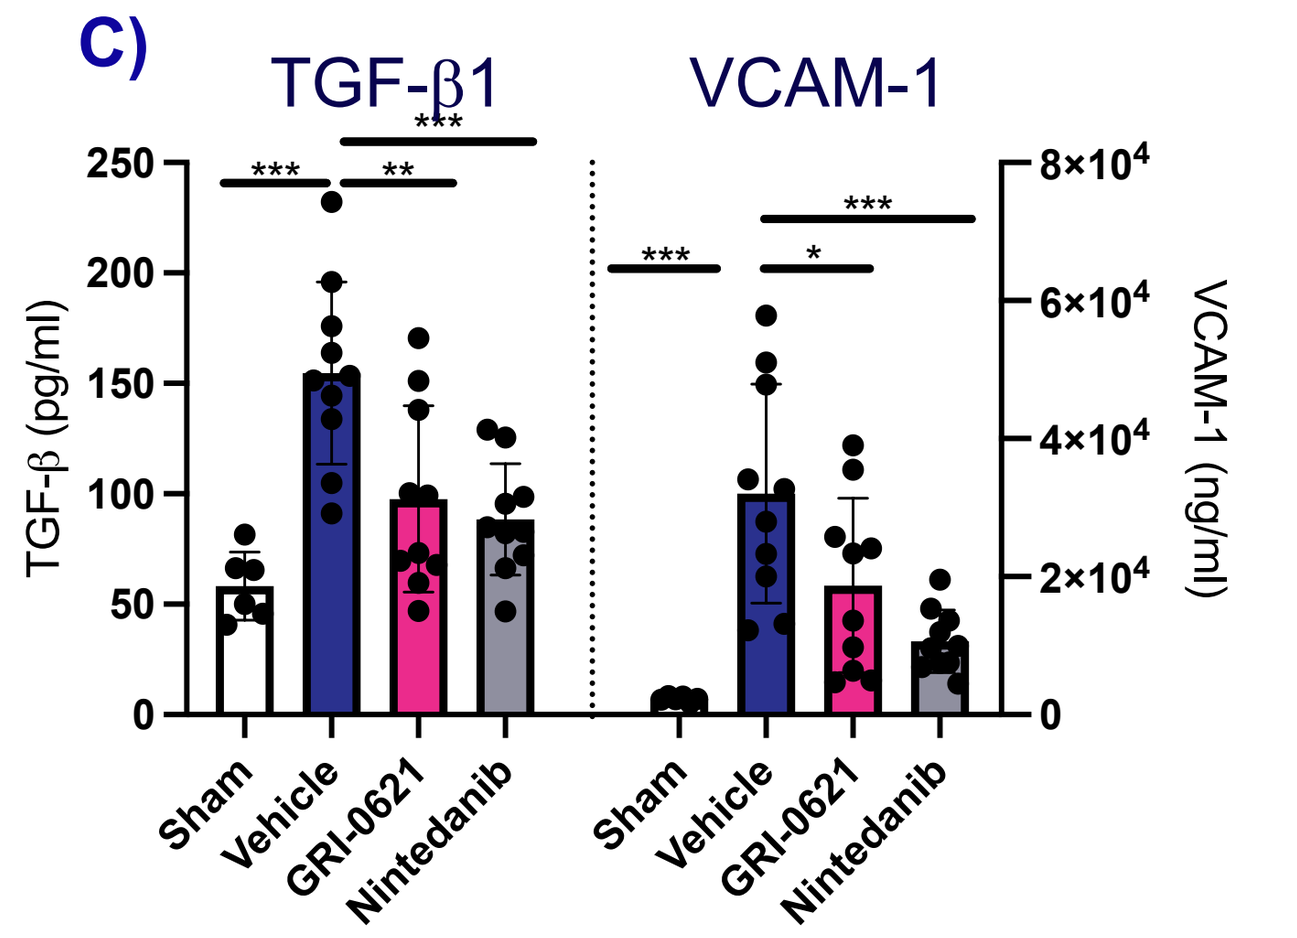
C)
TGF-β1
VCAM-1
TGF-β (pg/ml)
VCAM-1 (ng/ml)
Sham
Vehicle
GRI-0621
Nintedanib

Figure 4. A) Ashcroft scores in mice. B) Total cell counts and numbers of neutrophils in lung. C) Levels of TGF-β and VCAM-1 in lung homogenates. n = 10-12 per group, pooled from two independent experiments. Data presented as mean ± SEM. * p<0.05, ** p<0.01, *** p<0.005, **** p<0.001..

NCT06331624

GRI-0621 is being assessed in a 12-week, double-blind, randomized, placebo-controlled study in patients with IPF. The objective is to assess the safety and tolerability of GRI-0621 in the IPF patient population. In addition, the effect of GRI-0621 on a number biomarkers both from the blood and BAL will be evaluated. These include several biomarkers associated with disease progression (ProC3, ProC6) as well as NKT cell and other immune cell numbers and activity.

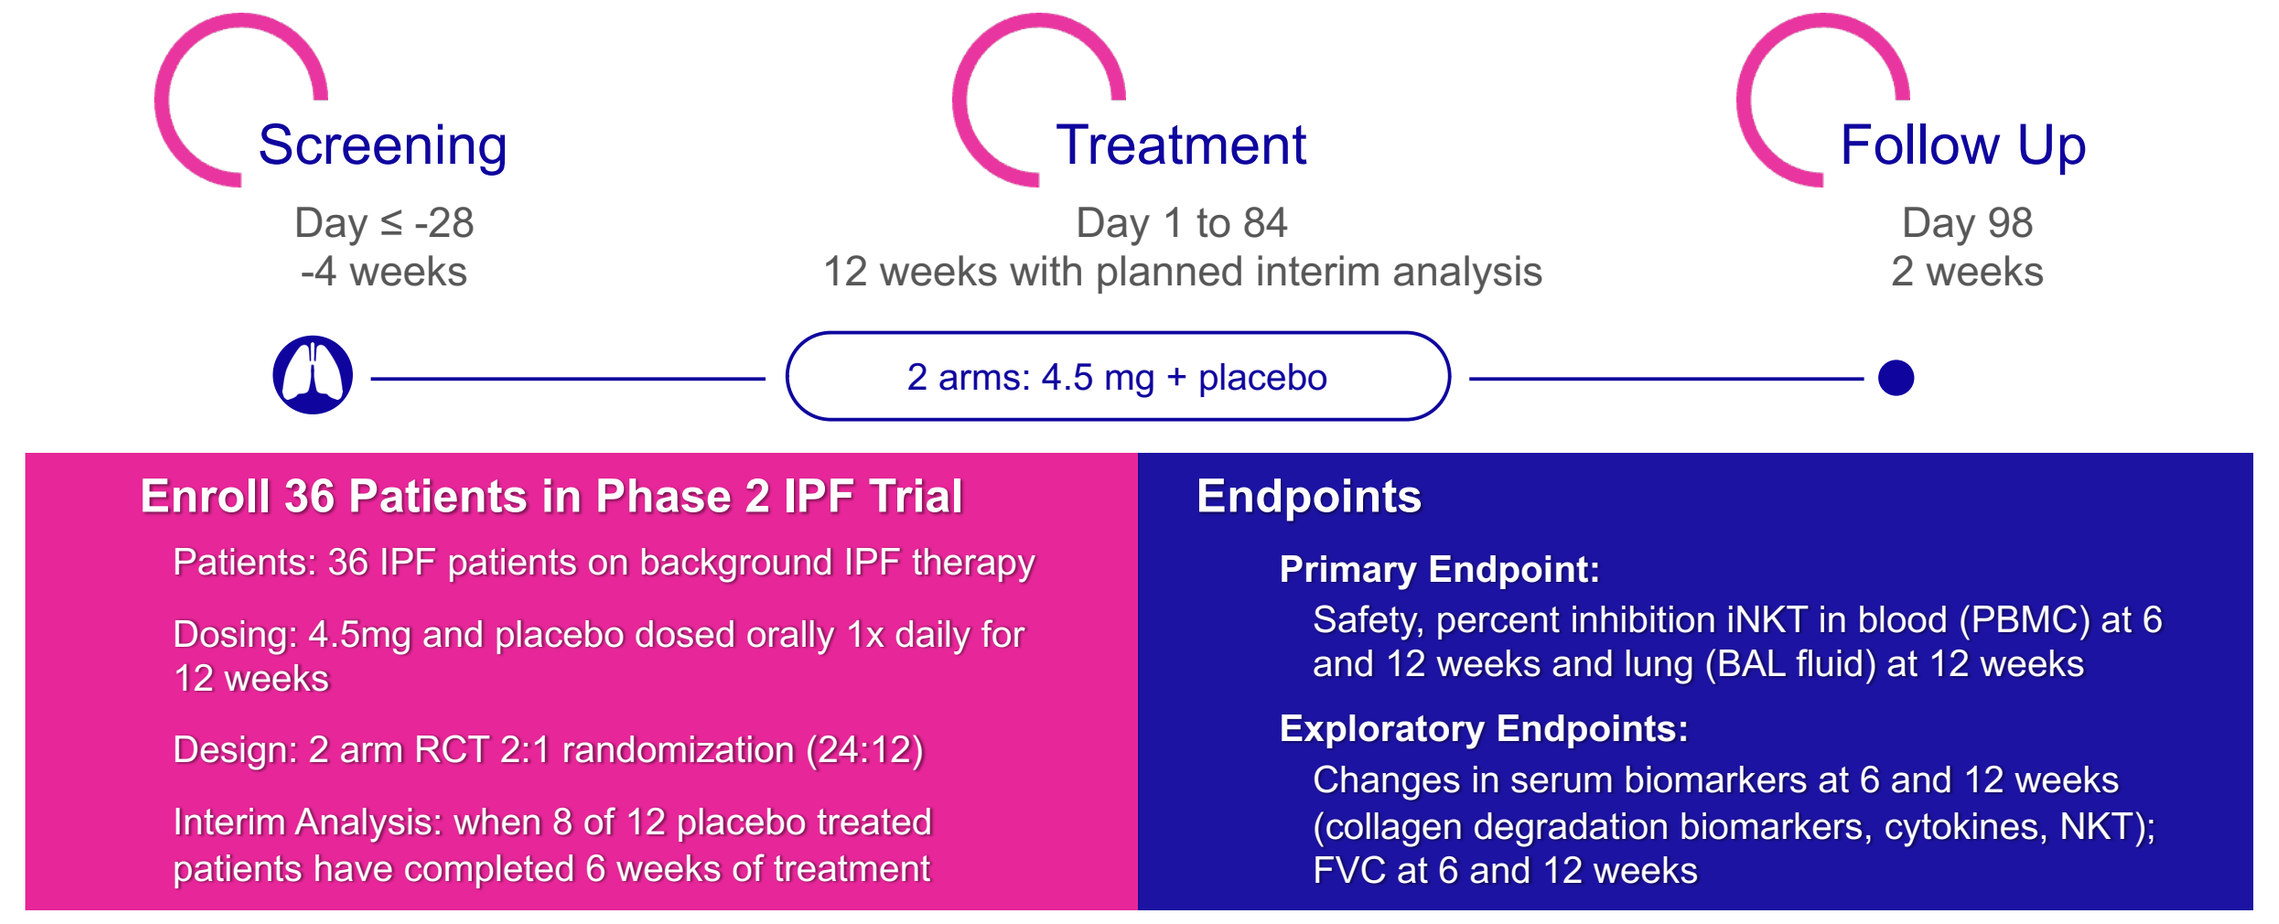
Screening
Day ≤ -28
-4 weeks
Treatment
Day 1 to 84
12 weeks with planned interim analysis
Follow Up
Day 98
2 weeks
2 arms: 4.5 mg + placebo
Enroll 36 Patients in Phase 2 IPF Trial
Patients: 36 IPF patients on background IPF therapy
Dosing: 4.5mg and placebo dosed orally 1x daily for 12 weeks
Design: 2 arm RCT 2:1 randomization (24:12)
Interim Analysis: when 8 of 12 placebo treated patients have completed 6 weeks of treatment
Endpoints
Primary Endpoint:
Safety, percent inhibition iNKT in blood (PBMC) at 6 and 12 weeks and lung (BAL fluid) at 12 weeks
Exploratory Endpoints:
Changes in serum biomarkers at 6 and 12 weeks (collagen degradation biomarkers, cytokines, NKT); FVC at 6 and 12 weeks

Conclusions

- iNKT cells are found in increased number and activity in the BAL of patients with IPF as compared to age matched controls
- iNKT cells are an early initiator of fibrotic disease effecting numerous pathways leading to fibrosis and inflammation
- GRI-0621 is a small molecule inhibitor of iNKT cell activity facilitated by the binding to RAR receptors on iNKT cells
- In an animal model of IPF, GRI-0621 was shown to reduce the fibrotic score, inhibit the production of TGFβ and VCAM-1 and reduce the immune cell infiltrate similar to the active comparator, nintedanib
- GRI-0621 is presently being evaluated in a Phase 2a biomarker study in patients with IPF with interim data available in Q3 2024 and top-line data available in Q4 2024

References

1. Kumar V, Hertz M, Agro A, Byrne AJ. Type 1 invariant natural killer T cells in chronic inflammation and tissue fibrosis. Front Immunol 2023;14:1260503.
2. Wehr A, Baeck C, Heymann F, Niemietz PM, Hammerich L, Martin C, *et al.* Chemokine Receptor CXCR6-Dependent Hepatic NK T Cell Accumulation Promotes Inflammation and Liver Fibrosis. *The Journal of Immunology* 2013;190:5226–5236.
3. Maricic I, Marrero I, Eguchi A, Nakamura R, Johnson CD, Dasgupta S, *et al.* Differential Activation of Hepatic Invariant NKT Cell Subsets Plays a Key Role in Progression of Nonalcoholic Steatohepatitis. *The Journal of Immunology* 2018;201:3017–3035.
4. Maricic I, Sheng H, Marrero I, Seki E, Kisseleva T, Chaturvedi S, *et al.* Inhibition of type I natural killer T cells by retinoids or following sulfatide-mediated activation of type II natural killer T cells attenuates alcoholic liver disease in mice. *Hepatology* 2015;61:1357–69.
5. Stegmann KA, Robertson F, Hansi N, Gill U, Pallant C, Christophides T, *et al.* CXCR6 marks a novel subset of T-bet(lo)Eomes(hi) natural killer cells residing in human liver. *Sci Rep* 2016;6:26157.

GRI Bio, Inc. © 2024

Drs. Agro, Kumar, and Hertz hold financial interest in GRI Bio, Inc. Commercial support provided by GRI Bio, Inc.